Texture profile analysis, a valuable tool for pharmaceutical oral dosage form evaluation: case study on chewable softgel capsules

Quemeneur Fanny [1,2]; Igonin Annabel [1]; Herry Catherine [1]; Girod Fullana Sophie[2]

[1] CIRIMAT, Université Toulouse 3 Paul Sabatier, INP, CNRS (France)

[2] NextPharma, Ploërmel (France) Contact information: fanny.quemeneur@univ-tlse3.fr & annabel.igonin@nextpharma.com

PURPOSE

New concept of **Softgel Capsules (SGC)** has emerged: **chewable** SGC, which could permit elderly and pediatric populations to benefit from lipid-based formulations [1]. In a previous study, two ways have been explored to obtain chewable capsule shells made of gelatin (G):
by **enhancing plasticizer (P) content** (decreasing G/P ratio) and/or by **modifying polymer composition** (gelatin and starch blend).

In order to determine the relevance of these two formulation strategies, **Texture Profile Analysis (TPA),** a widely used method in food domain and more recently in pharmaceutical field[2], has been used in this study to discriminate **chewable formulations** and **non-chewable formulations**. The choice of suitable formulation lies on an **accurate evaluation of the mechanical behavior** and **resulting mouthfeel** of the dosage form. Chewable SGC should have appropriate texture for a pleasant mouthfeel.
Two prototypes (Chew1 & Chew2), optimized compositions obtained with a mixture Design of Experiments (DoE), were compared to a non-chewable SGC reference (REF) with a **G/P of 2.5**.
Chew1 was a **starch-free** prototype with a **G/P of 0.81** and Chew2 contained **9% w/w of starch** and a **G/P of 0.67**.

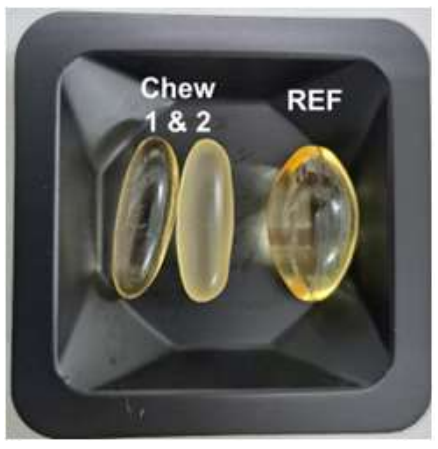
Chew
1 & 2
REF

Figure 1 : Visual aspect of tested SGC

MATERIAL AND METHODS

Materials: Capsule shells were made of pharmaceutical grade : 170B bovine hide gelatin (PB Leiner), glycerin (Cooper) and purified water. Starch was acid-modified and fluidized thin boiling starch, Cleargum® MB45 (Roquette). Capsules were filled with Medium Chain Triglycerides (MCT).

Softgel capsule manufacturing: Gel mass was prepared by pouring solid excipients to liquid excipients, performing the melting step at 80°C under stirring, then vacuuming them to obtain a bubble-free gel mass.
The reference capsule (REF) was an Oval #10 capsules, filled with 500mg MCT.
Chew 1 & 2 were Oval #10 capsules, filled with 250 mg MCT.
Ribbon thickness were set at 35 milli inch.
SGC were manufactured using a small-scale machine (Bochang, Korea) (Fig. 2), then dried on trays at ~20% RH conditions at room temperature.

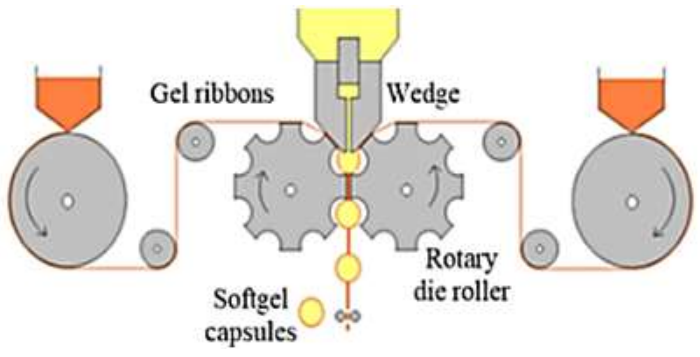
Gel ribbons
Wedge
Rotary
die roller
Softgel
capsules

Figure 2 : Rotary Die Technology for SGC manufacturing

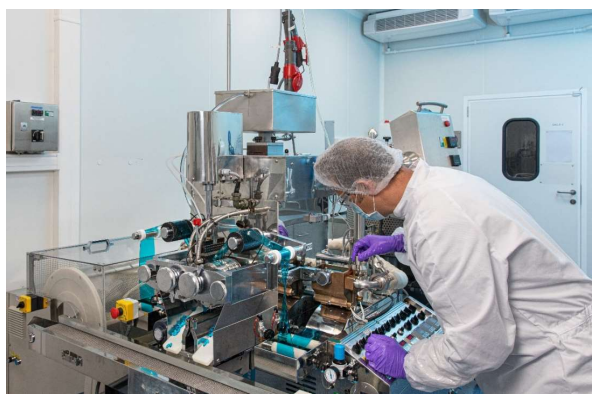

Texture profile analysis: Double compression test was performed on dried capsules (Fig. 3), using **TA-XT plus (Stable Micro Systems)** with an acrylic probe (P/20P) [3].

TPA experiment was performed on five units per capsule type (n=5).
The cylindrical probe compressed the SGC, laid along the sealing, up to 30% of strain with a trigger force of 0.05N, at a constant speed of 5 mm/min with a time elapsed of 5s between the two compression events.

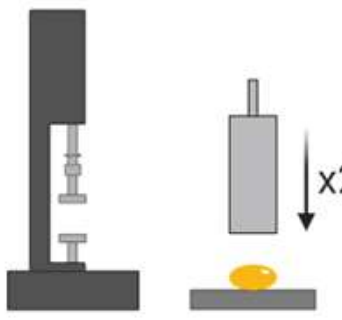
x2

Figure 3 : Texturometer set up

Four texture parameters were considered as key parameters to evaluate chewable properties:
a **minimized hardness**, linked to an easy bite of the capsules with minimal jaw force. Hardness values are obtained by direct reading of max force values;
a **minimized chewiness,** corresponding to the force required to chew a solid into a state suitable to swallow (hardness * springiness *cohesiveness);
a **maximized resilience and springiness,** linked to the gummy-like texture with elastic recovery, of the first deformation for resilience and during the time that elapses between the two compression events for springiness.

$$\text{Resilience} = \frac{\text{withdrawal area 2:3}}{\text{compression area 1:2}} \qquad \text{Springiness} = \frac{\text{duration of 2}^{\text{nd}}\text{ compression 4:5}}{\text{duration of 1}^{\text{st}}\text{ compression 1:2}}$$

RESULTS

Softgel capsule manufacturing

Capsules were underfilled, for the purpose of maximizing the evaluation of the shell texture and properties, in comparison to usual filling volume.
The capsules produced presented an adequate shape with a good sealing ratio under standard operating conditions and no leaking capsules were observed for the two prototypes.
Chew2 capsules appeared easier to handle than Chew1. This could be attributed to their surface rugosity, linked to the presence of starch granules in the continuous gelatin matrix[4], reducing capsule stickiness and giving them a frosted aspect while starch-free formulations (Chew1 and REF) are clear capsules (Fig. 1).

Texture profile analysis

TPA profiles and related results are detailed in Fig. 4 and Table 1.
Suitable chewable SGC should present an easy rupture with a soft gummy-like elastic texture.

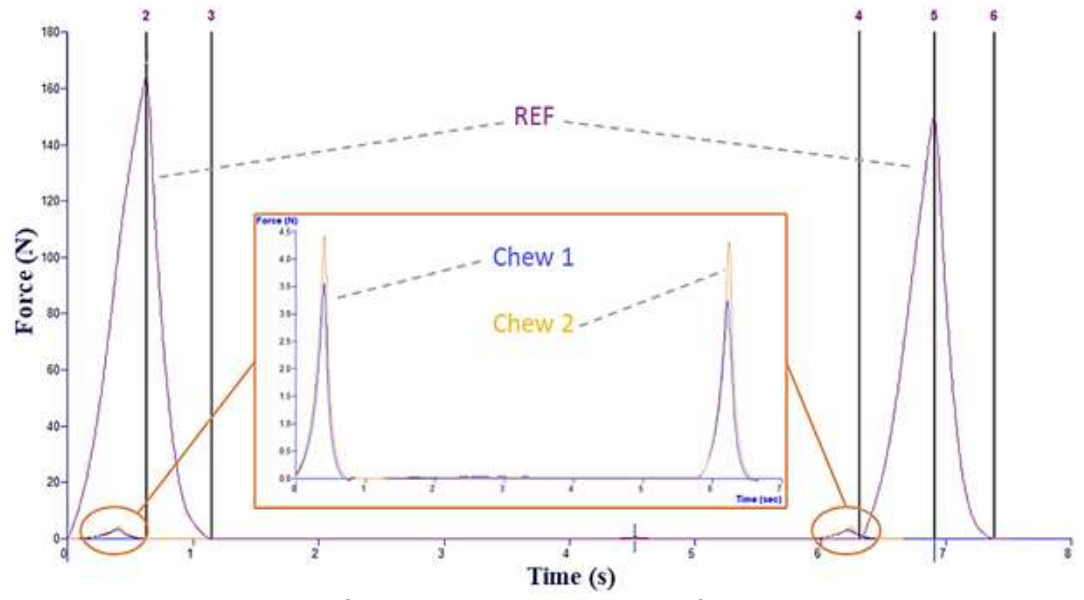
REF
Chew 1
Chew 2
Force (N)
Time (s)

Figure 4 : Texture Profile Analysis, Force by time plot of two compression cycles

Chew1 and Chew2 were **~40 times softer** (Hardness values are reduced) and much **easier to chew** (~30 times) than conventional SGC (REF), due to their higher plasticizer content.

The prototypes also exhibited **higher elasticity** than the reference capsules as they showed higher resilience and springiness. Chewable prototypes showed similar texture with no significant differences.

Table 1 : Mechanical properties of tested SGC

#	Hardness (N) ± SD	Resilience (%) ± SD	Springiness (%) ± SD	Chewiness (N) ± SD
Chew1	3.66ᵃ ± 0,22	59.20ᵃ ± 1.28	98.55ᵃ ± 0.52	355ᵃ ± 28
Chew2	4.27ᵃ ± 0.25	63.29ᵃ ± 2.23	98.08ᵃ ± 0.65	424ᵃ ± 27
REF	164.63ᵇ ± 13.82	48.70ᵇ ± 4.32	91.17ᵇ ± 3.64	12160ᵇ ± 2192

Different uppercase letters in the same column indicate significant differences (One-way Anova with multiple Tuckey Test; p<0.05)

CONCLUSION

TPA can serve as a **valuable tool** for **effective comparison of softgel capsule dosage forms**. It supports the **application suitability** for the **development** of **chewable dosage forms** and permits to **discriminate chewable and non-chewable formulations**. Both developed SGC prototypes, characterized by a high plasticizer content and an optional starch addition, presented expected and **optimized chewable texture** (reduced hardness and chewiness, and maximized elastic recovery).

References

1. Feeney O.; Crum M.; McEvoy C.; Trevaskis N.; Williams H.; Pouton C.; Charman W.; Bergström C.; Porter C. 50 years of oral lipid-based formulations: Provenance, progress and future perspectives. Advanced Drug Delivery Reviews vol. 101 167–194 (2016).
2. Bogdan C.; Hales D.; Cornilă A.; Casian T.; Iovanov R.; Tomuță I.; Iurian S. Texture analysis – A versatile tool for pharmaceutical evaluation of solid oral dosage forms. International Journal of Pharmaceutics, 638. (2023).
3. Guiné R.; Correia P.; Reis C.; Florença S. Evaluation of texture in jelly gums incorporating berries and aromatic plants. Open Agric 5, 450–461 (2020).
4. Marfil P.; Anhê A.; Telis V. Texture and Microstructure of Gelatin/Corn Starch-Based Gummy Confections. Food Biophys 7, 236–243 (2012).

PBP world meeting – Wien – March 2024